End-user trainable automatic antibiotic-susceptibility testing by disc diffusion using machine vision

K. Dijkstra[1], M. Berntsen[2], J. van de Loosdrecht[1] and W. J. Jansen[1]

1) NHL University of Applied Sciences, Center of Expertise Computer Vision, Leeuwarden, The Netherlands. 2) BD, Drachten, The Netherlands.

Objectives

BD Kiestra provides a workflow where digital images of Petri-dishes used in antibiotic susceptibility testing by disc diffusion can be automatically analyzed using machine vision algorithms.

The objective of this study is to develop and test a system which automatically optimizes a zone measurement algorithm to yield results close to zone measurements of a human end-user.

Methods

The main design principle is that the lab technician does not have to know anything about machine vision to train the system. Technical configuration is handled by Artificial Intelligence.

Three digital image sets of Petri-dishes using different illuminations from two European microbial laboratories have been selected from the daily routine to test the system. Evaluation is performed using a two-fold cross-validation.

Results

Blue and red circles are manual and automatic measurements respectively.
Crossed circles are automatically rejected measurements.

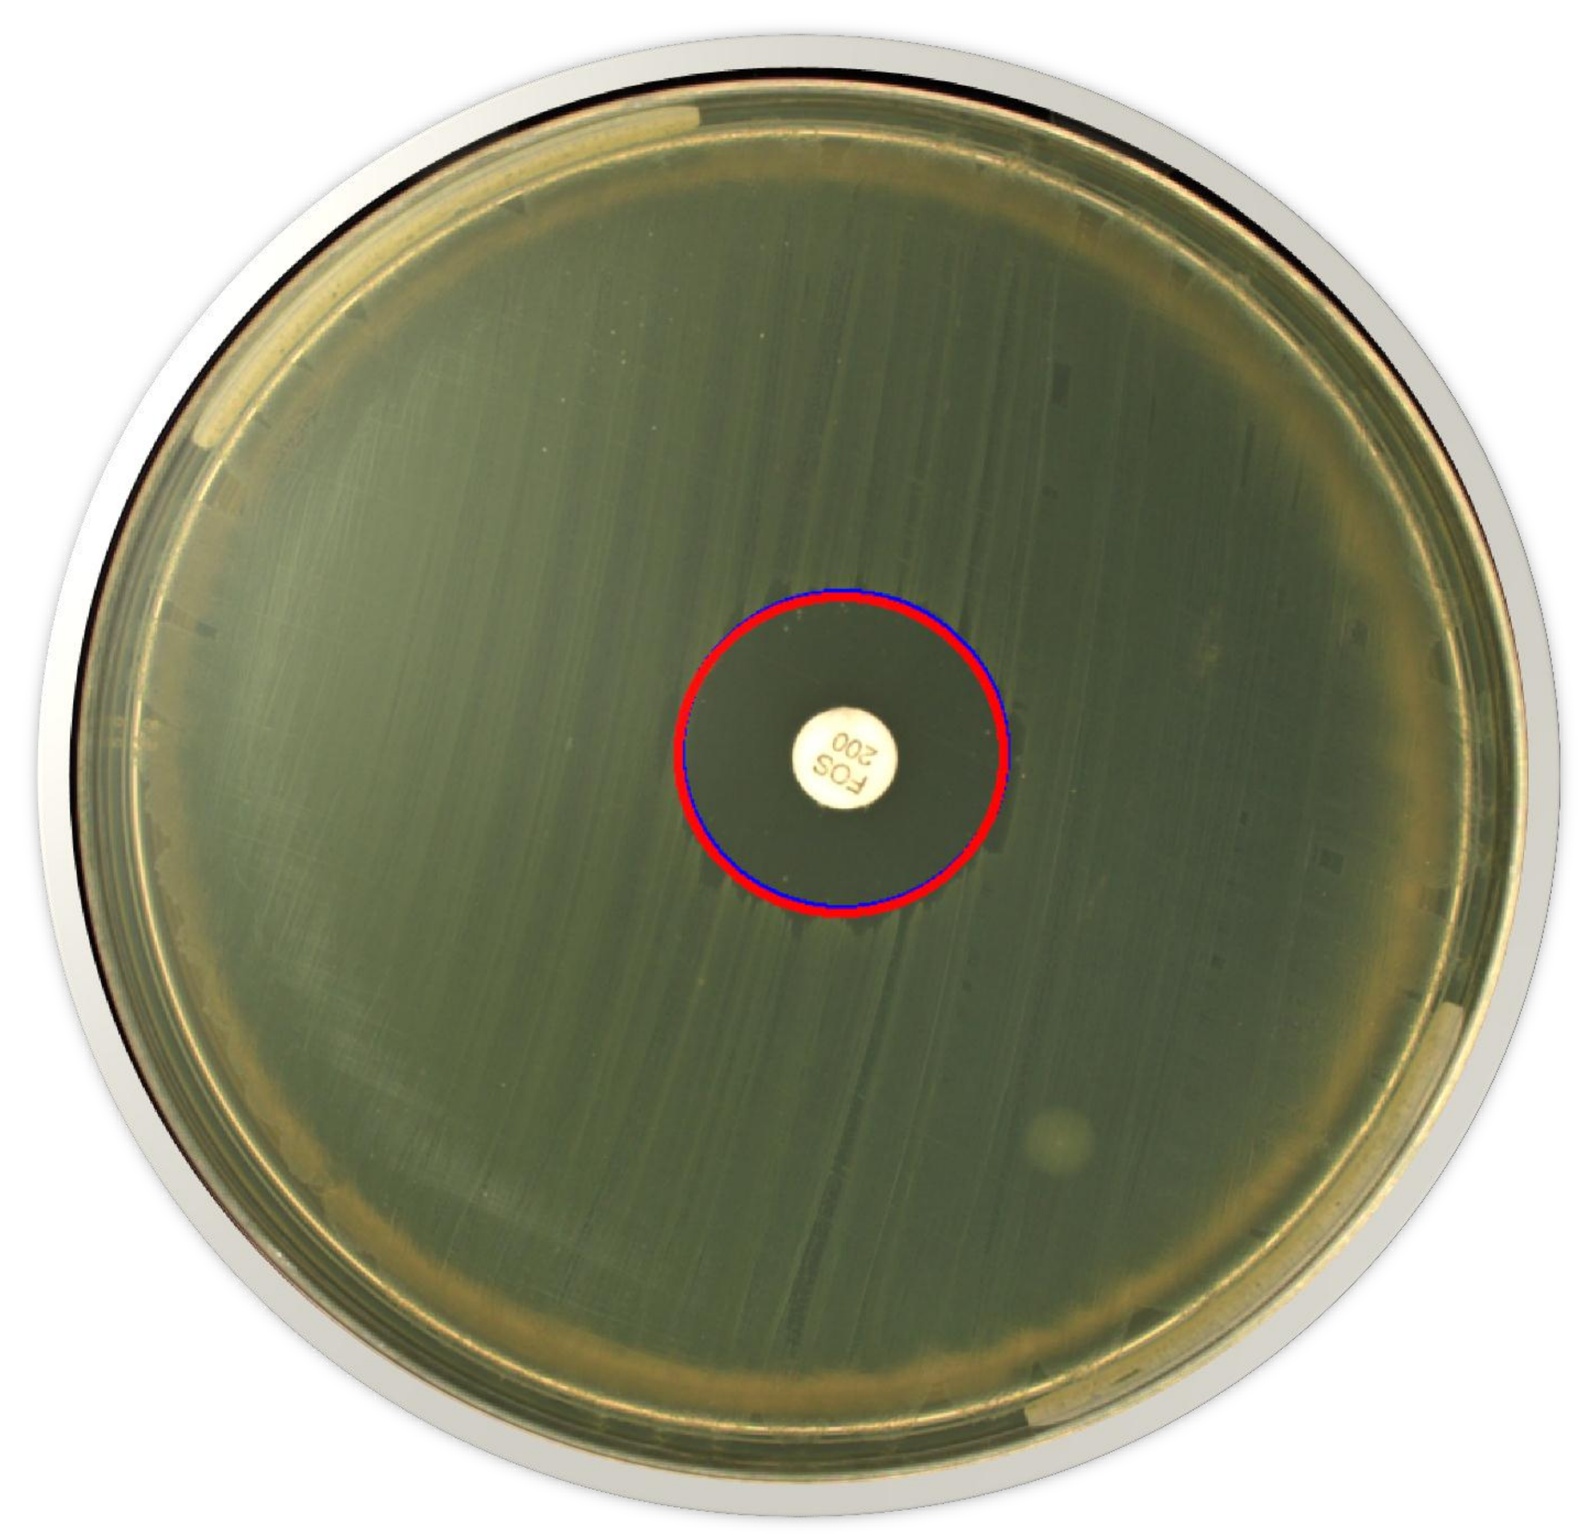

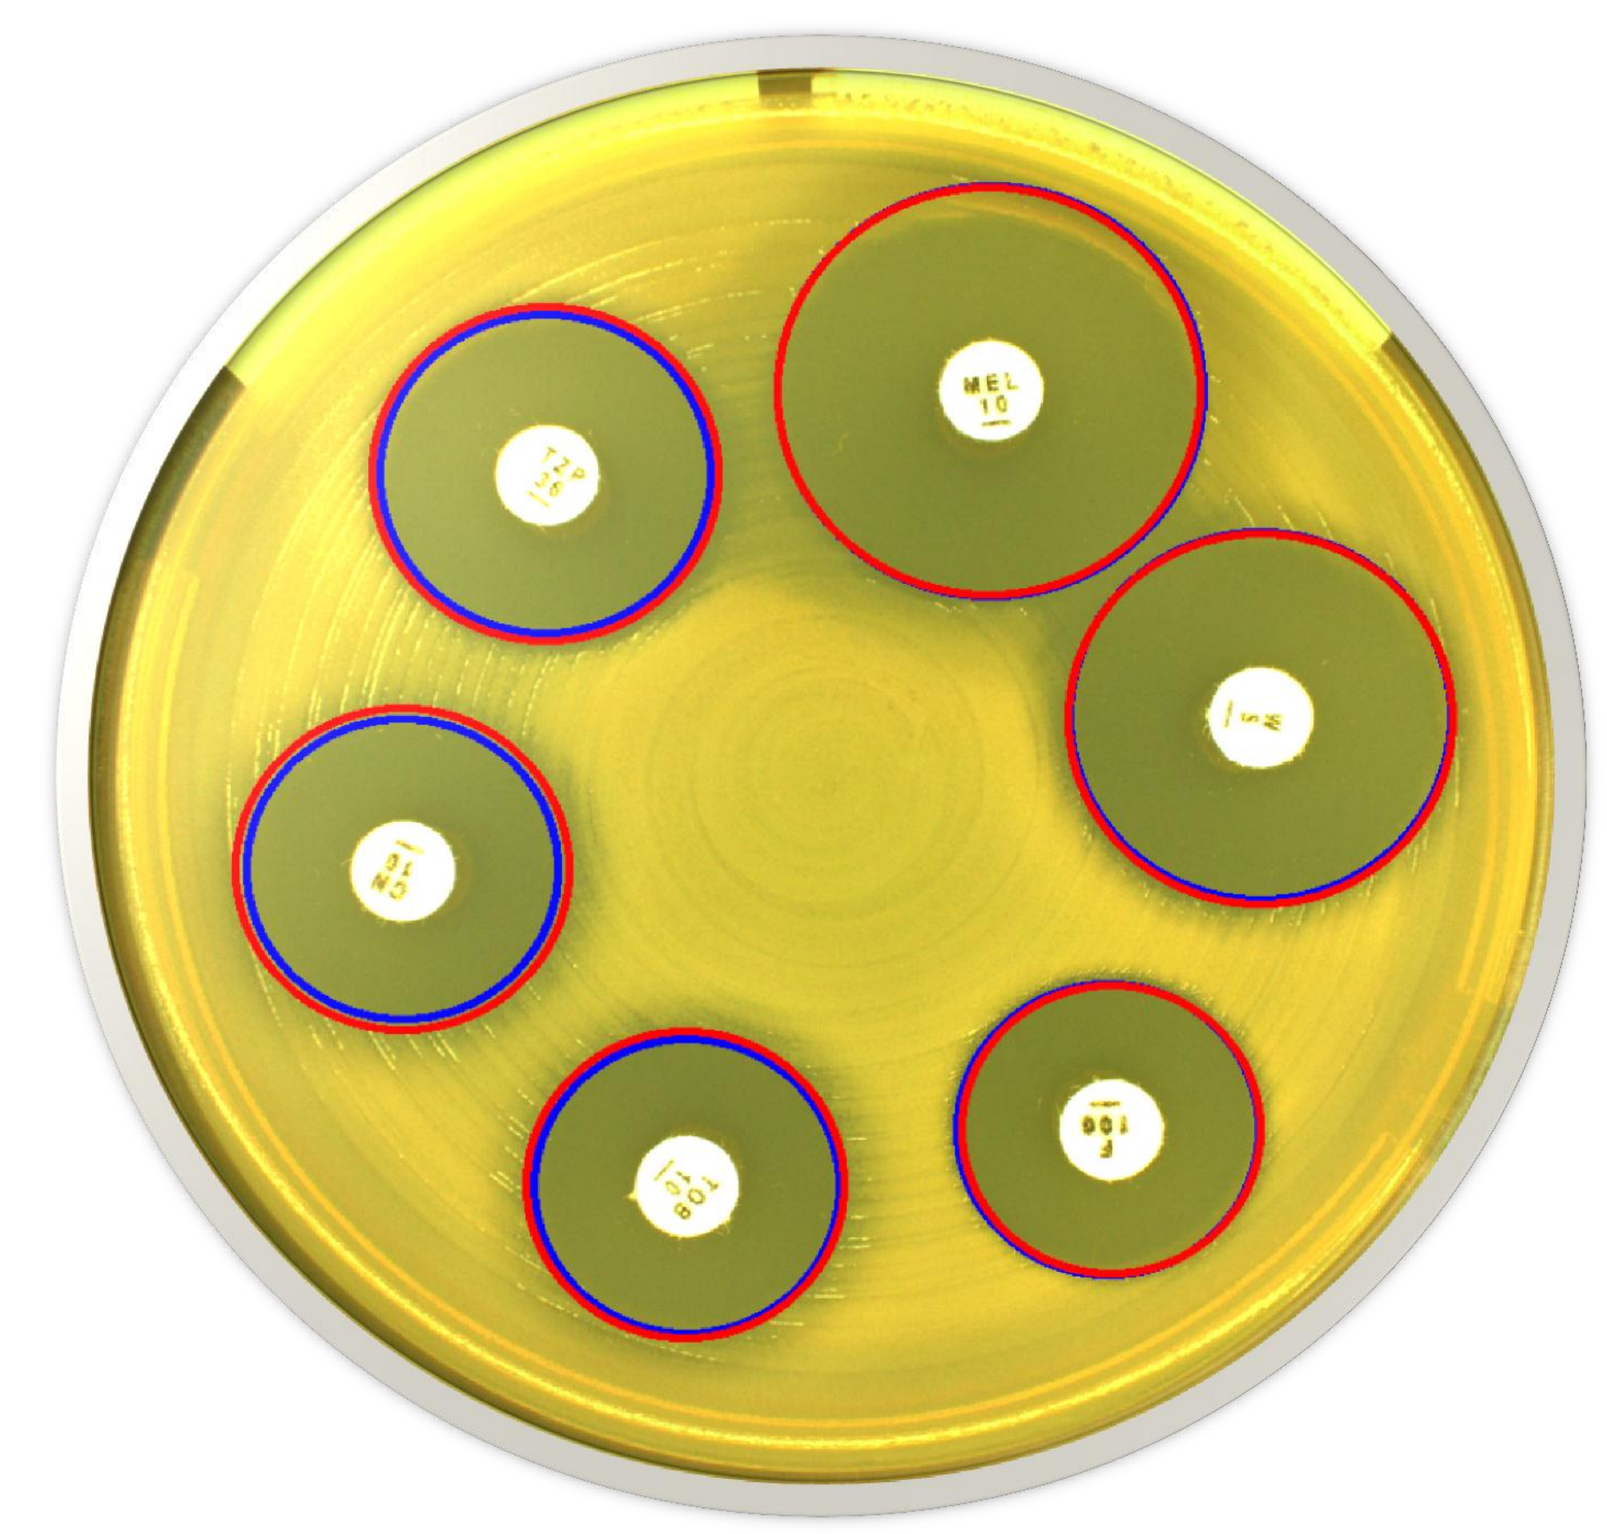

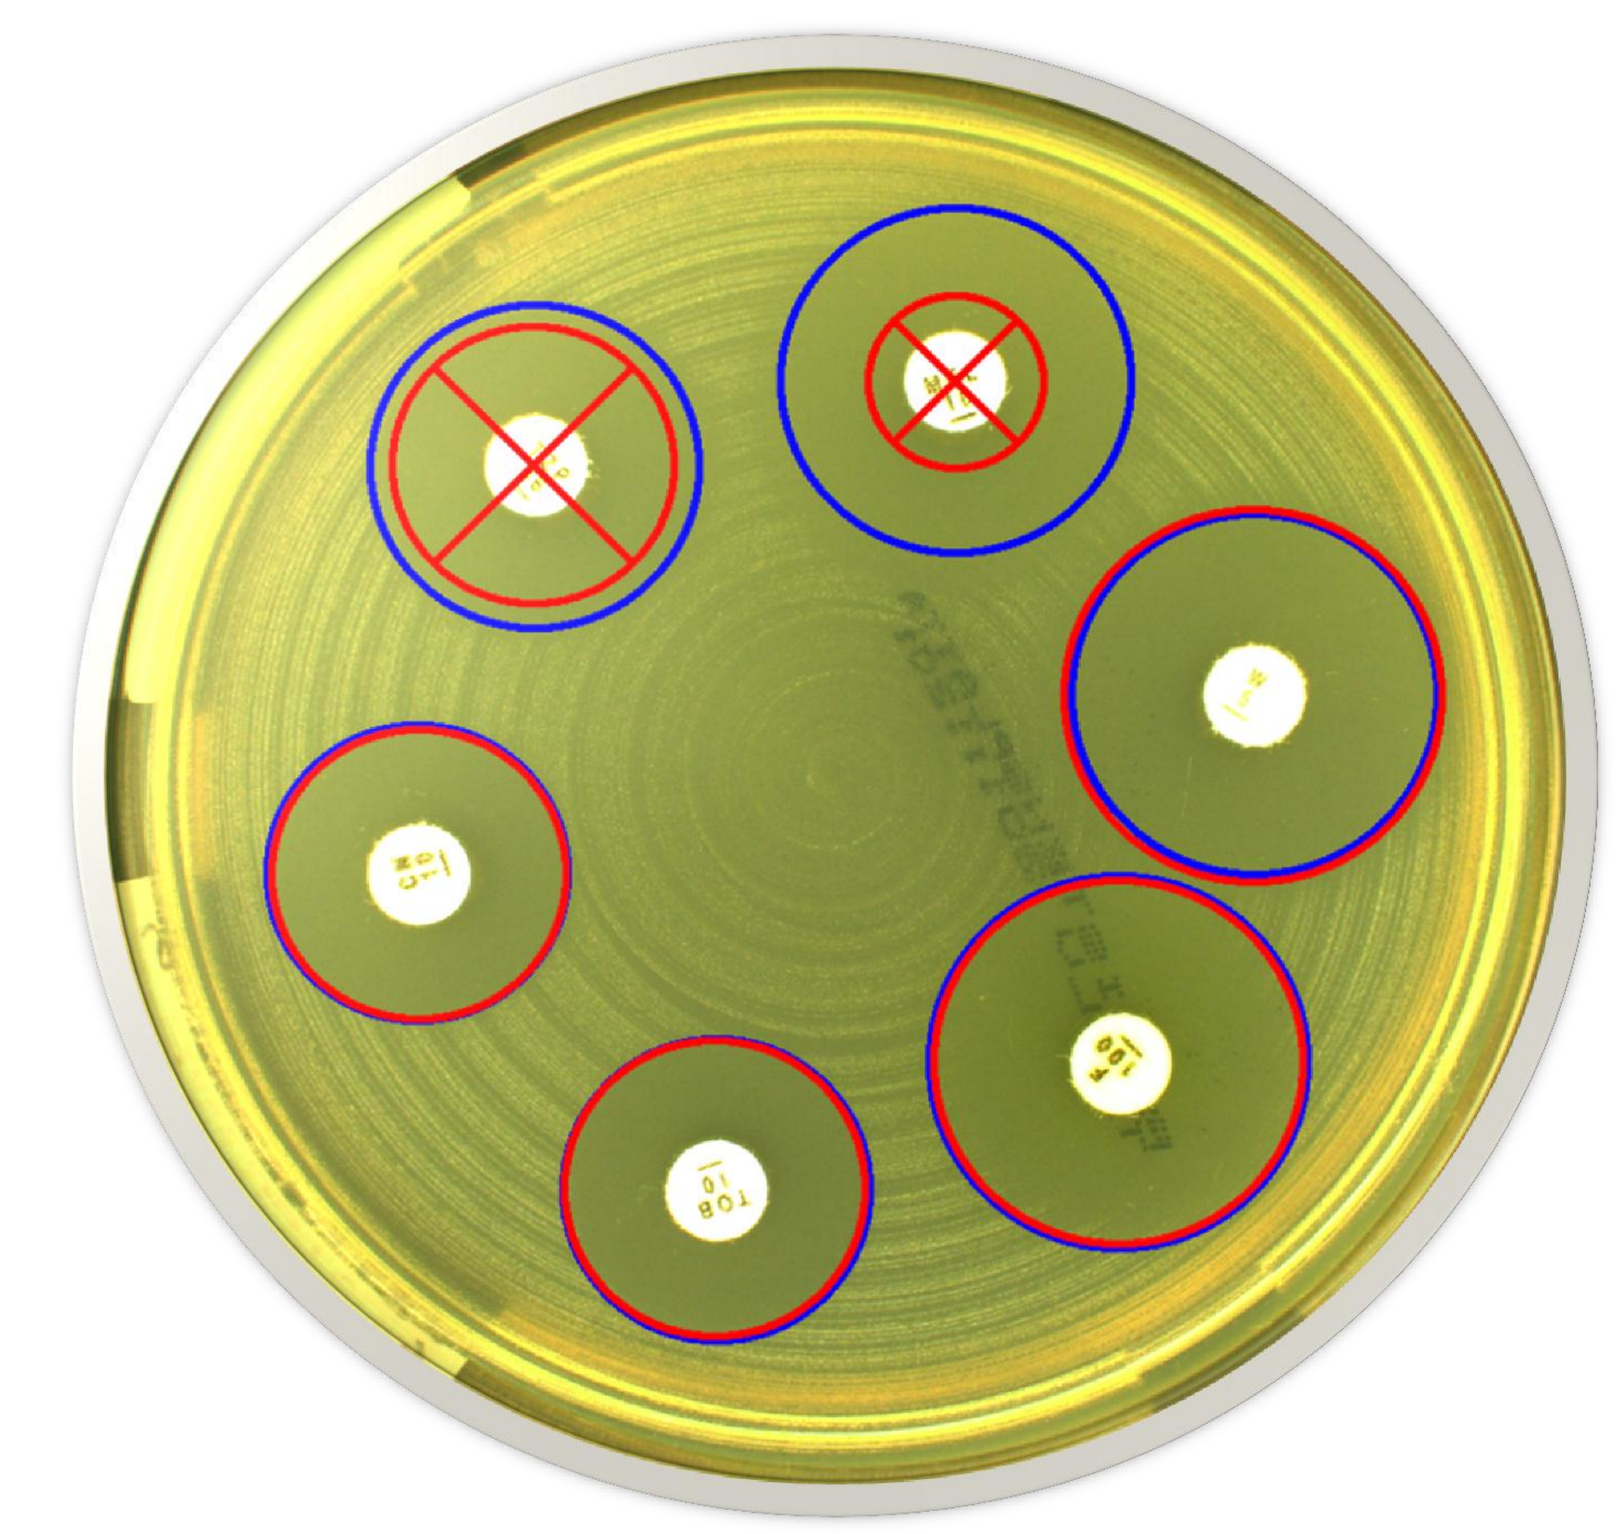

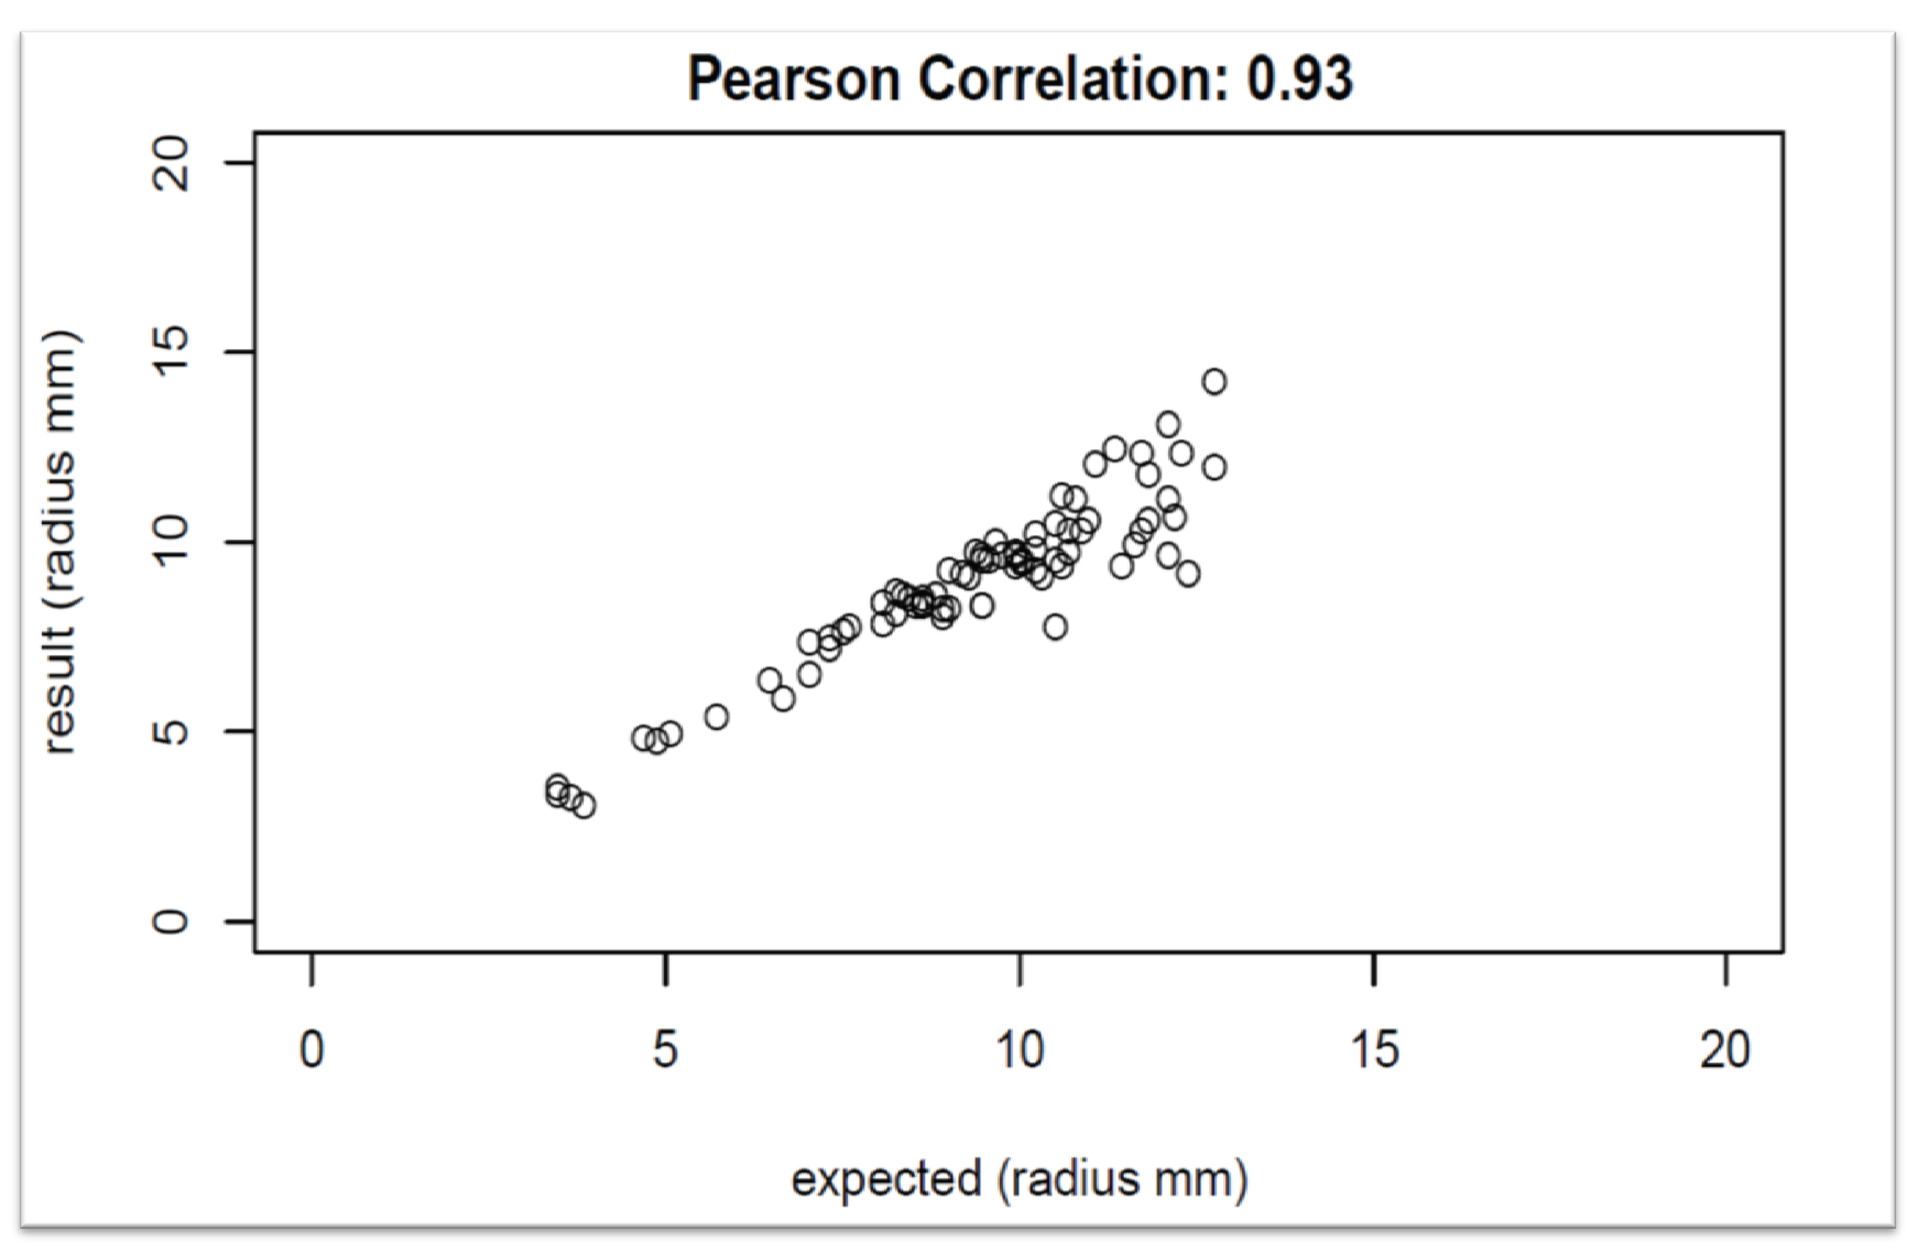
Pearson Correlation: 0.93
result (radius mm)
expected (radius mm)

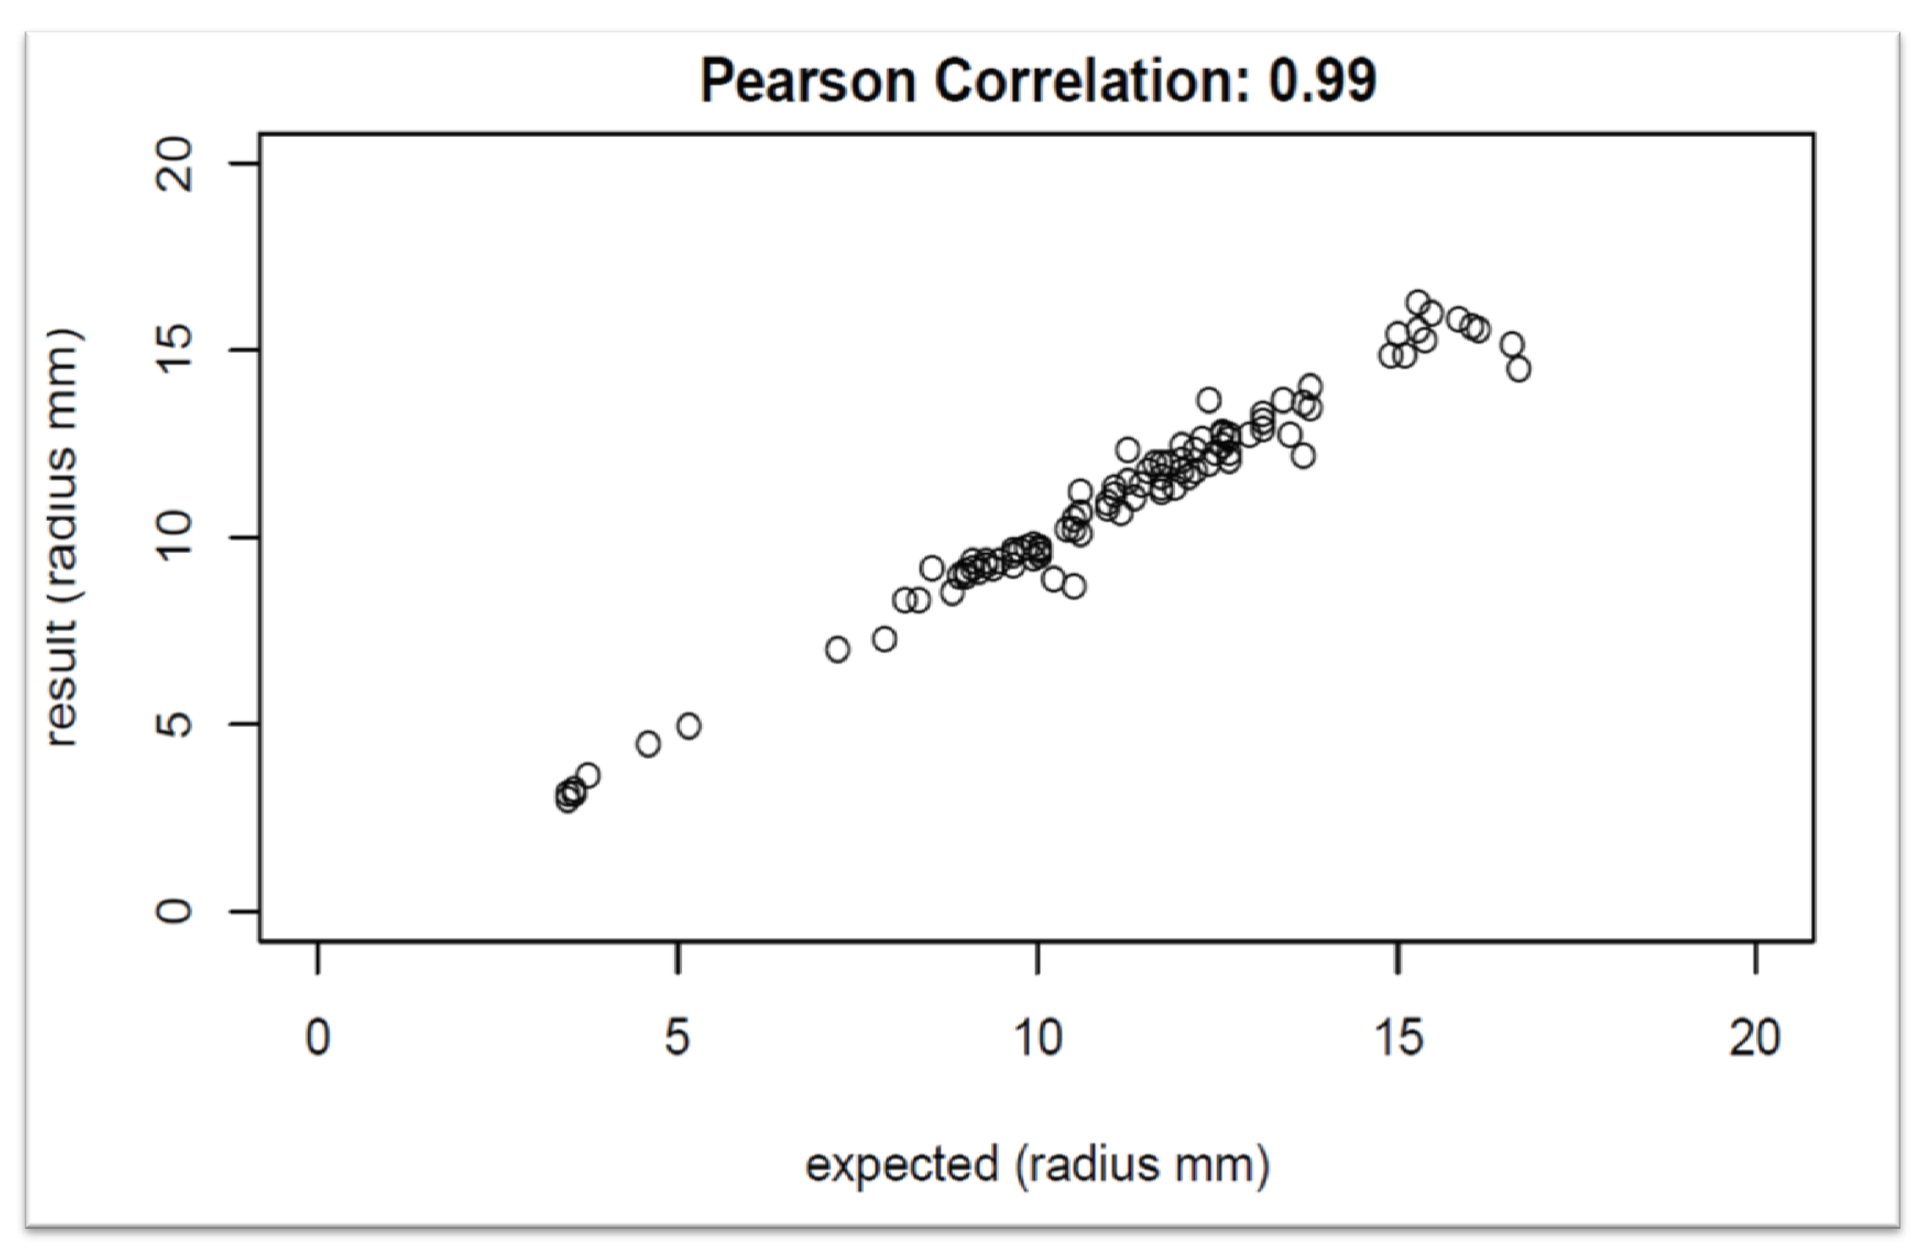
Pearson Correlation: 0.99
result (radius mm)
expected (radius mm)

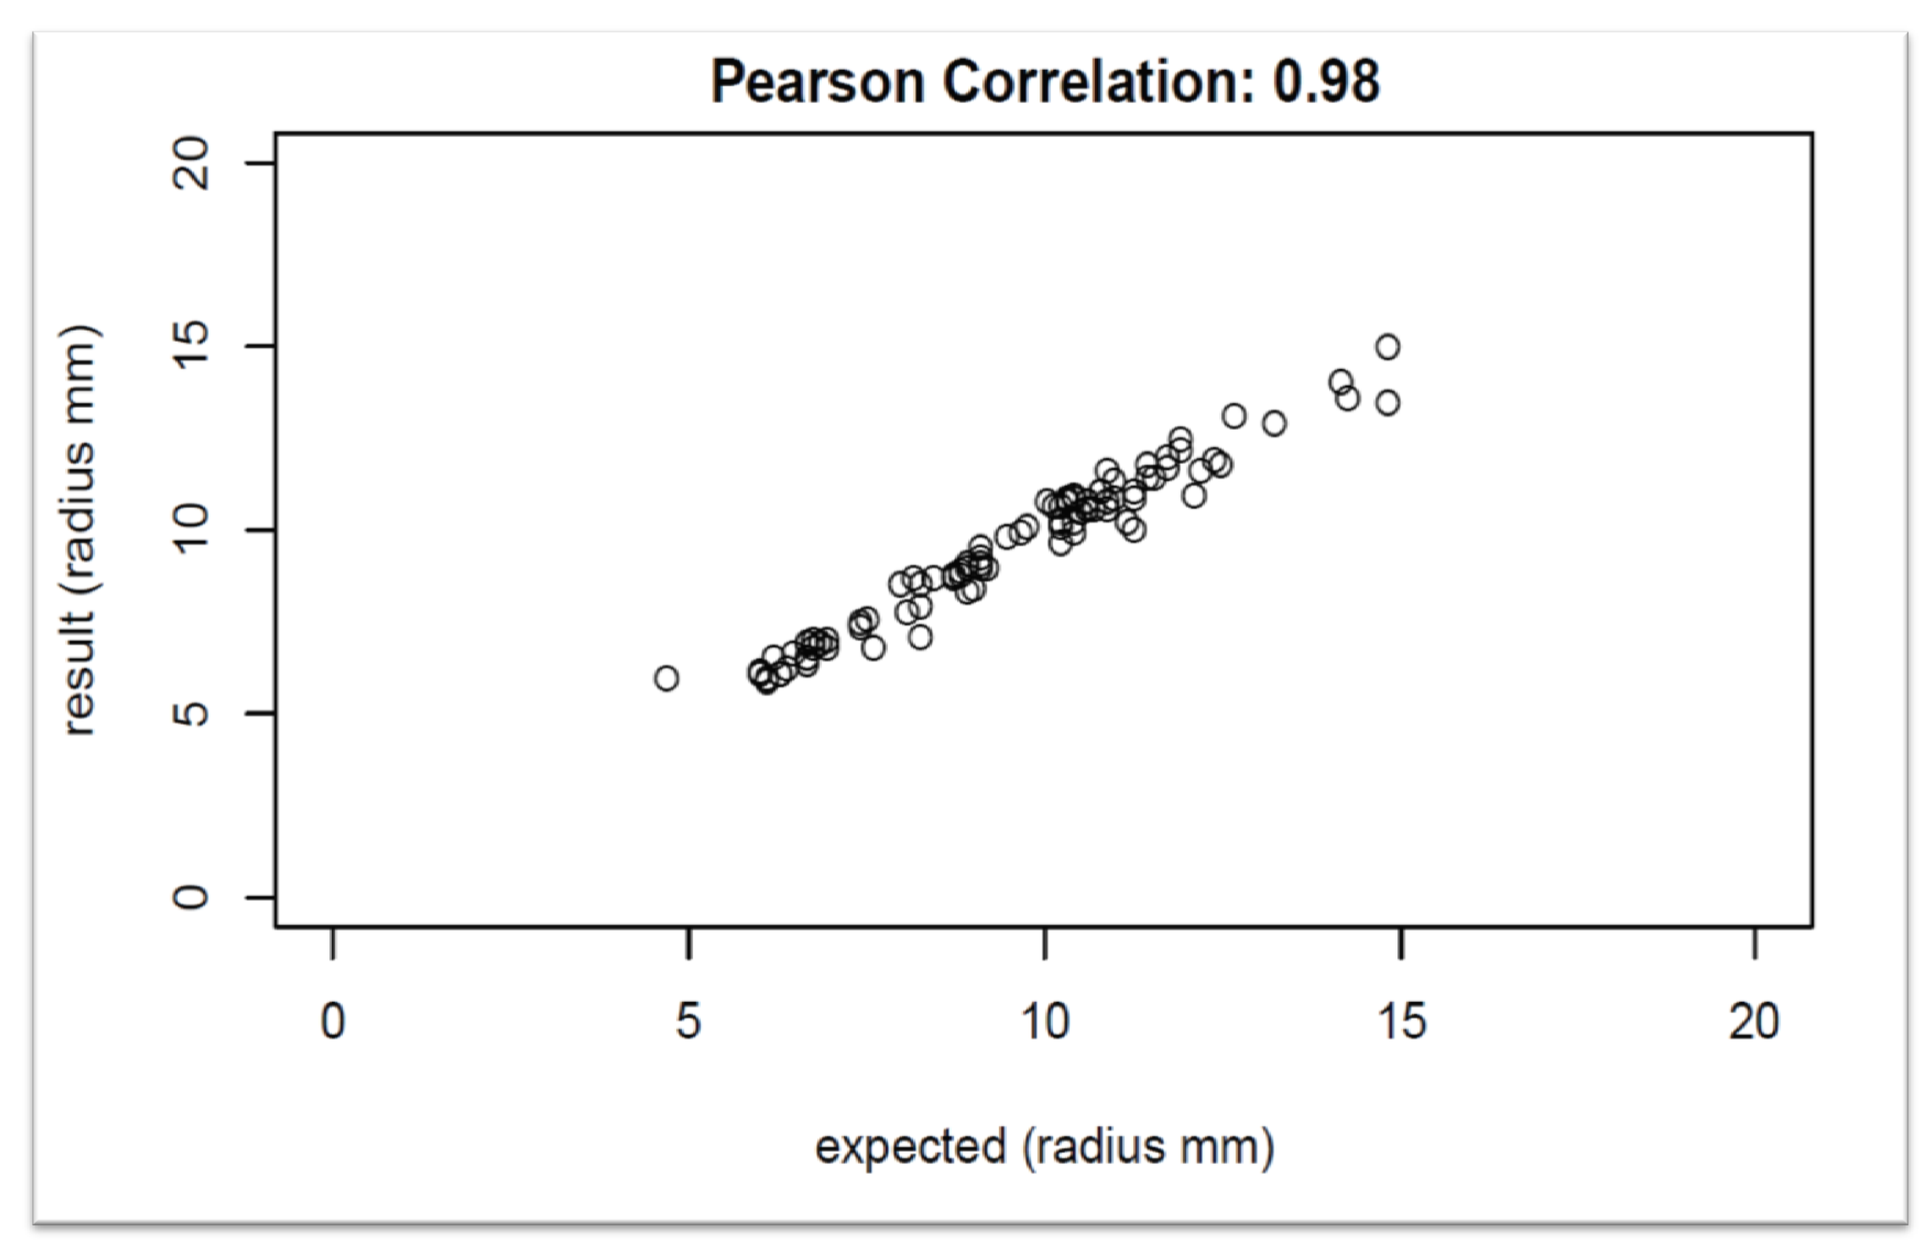
Pearson Correlation: 0.98
result (radius mm)
expected (radius mm)

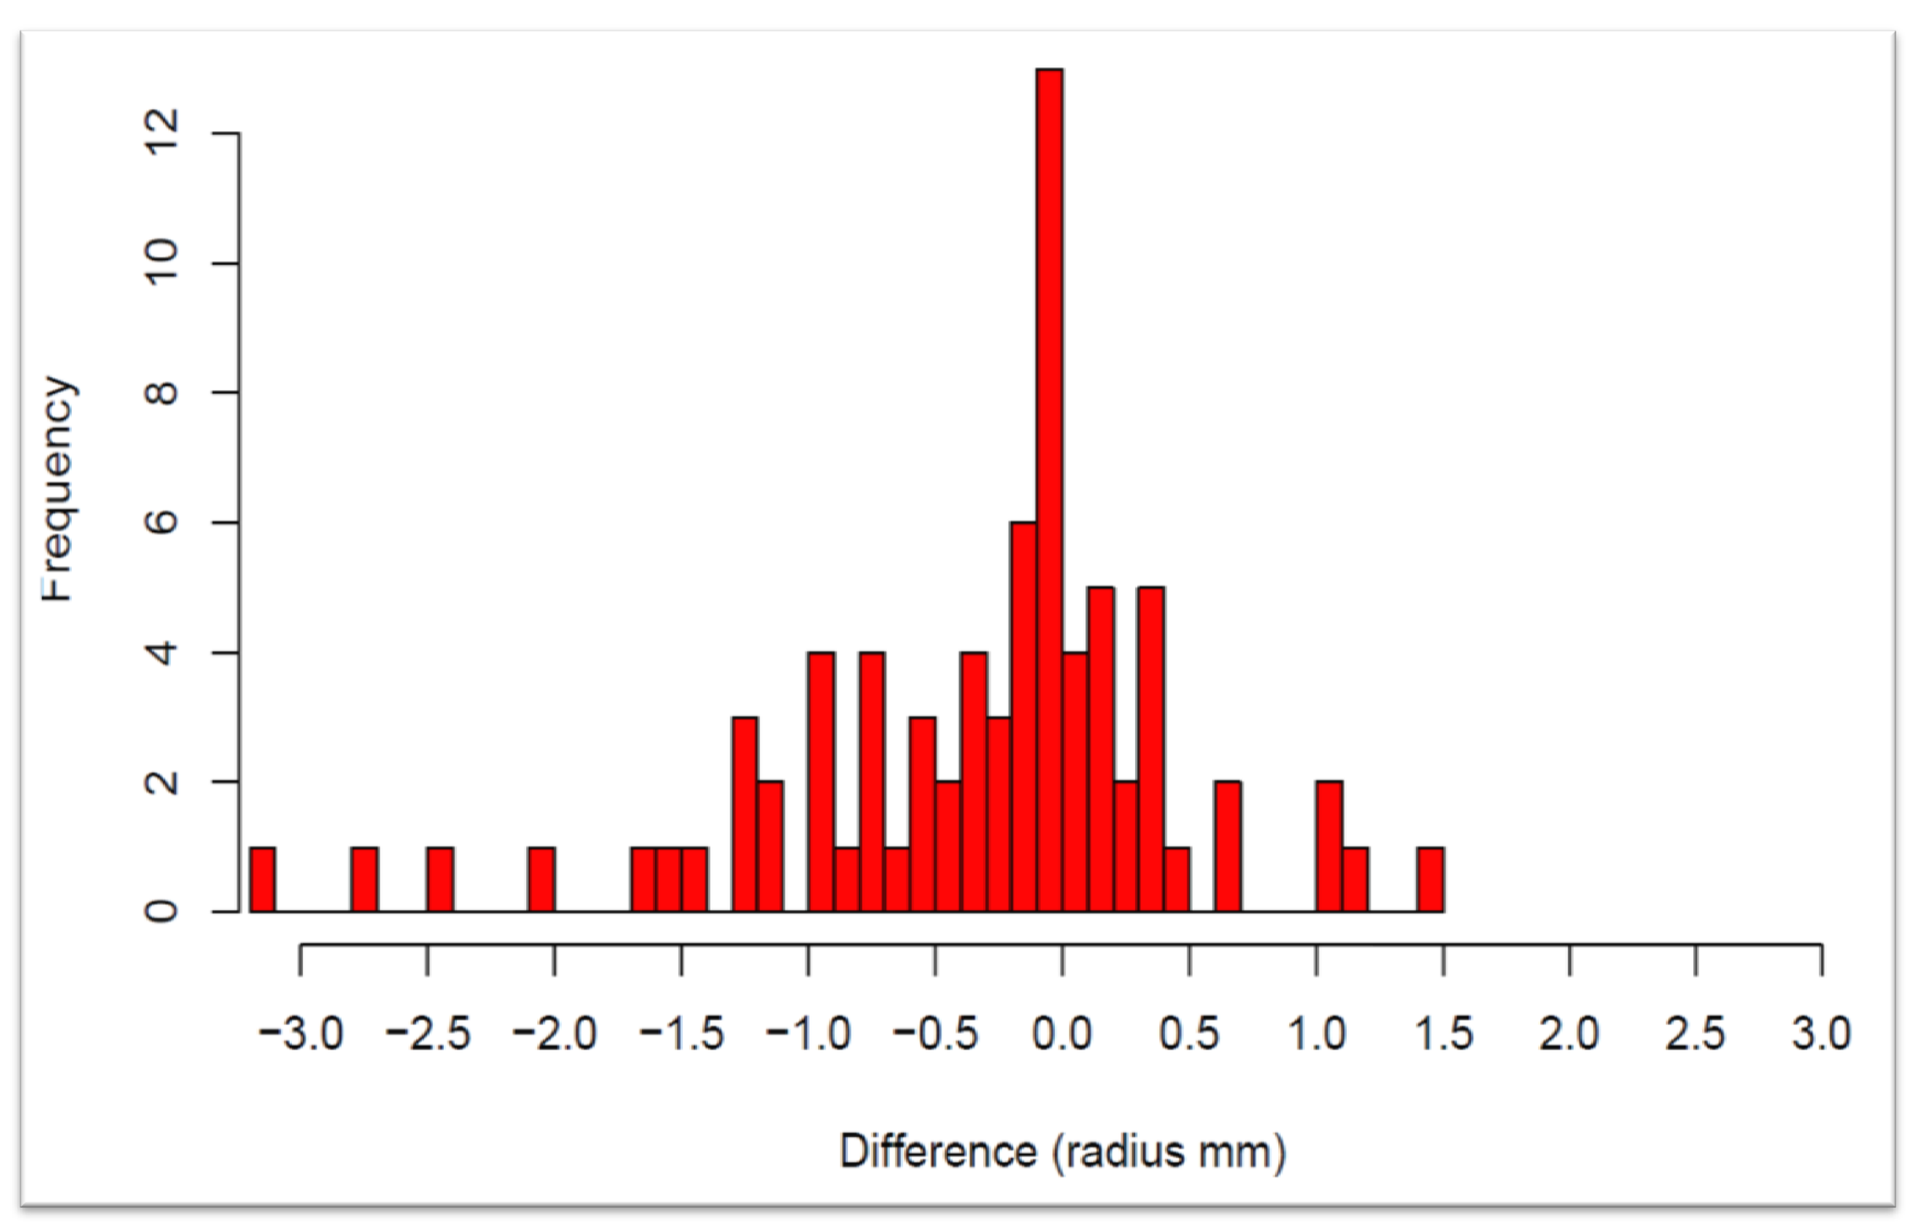
Frequency
Difference (radius mm)

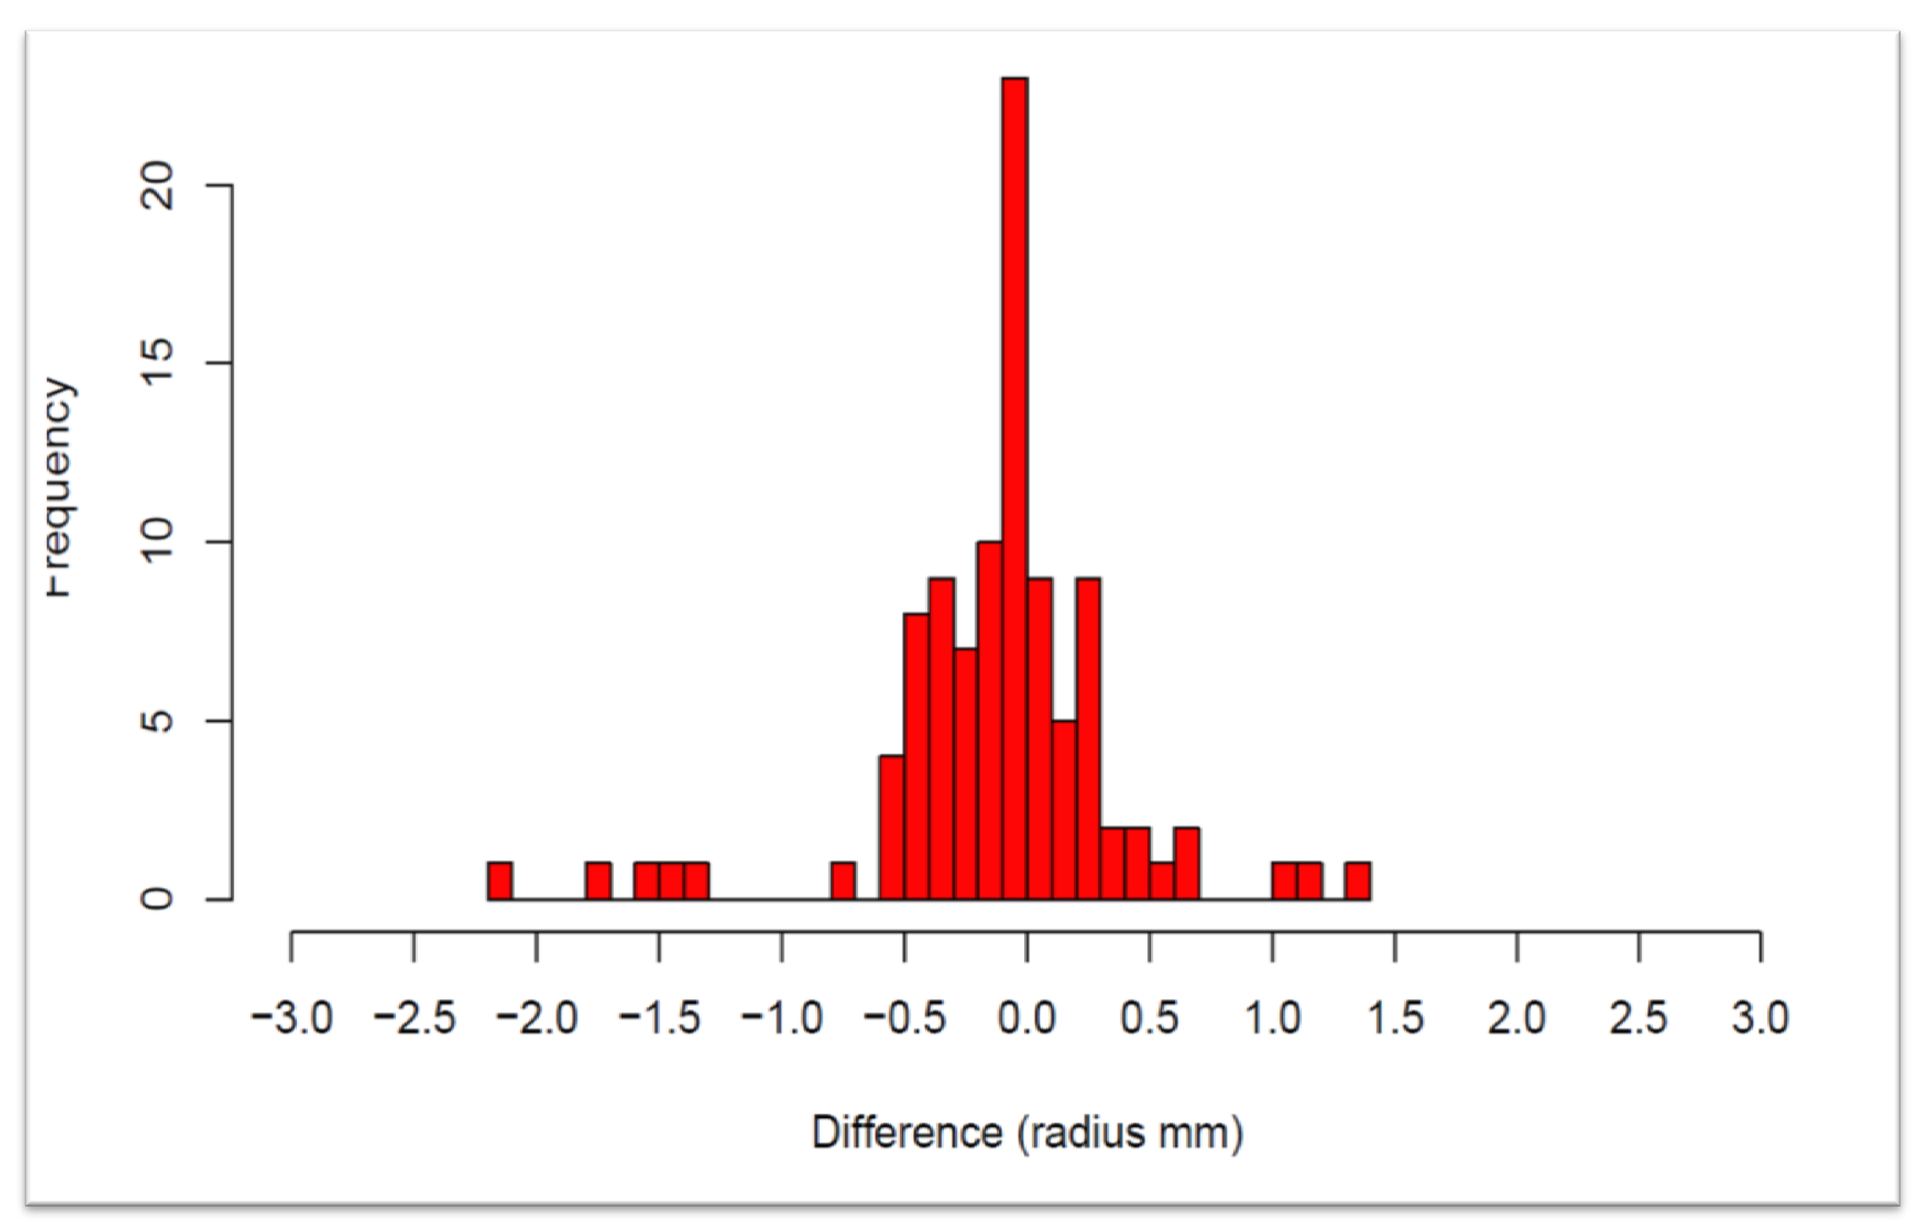
Frequency
Difference (radius mm)

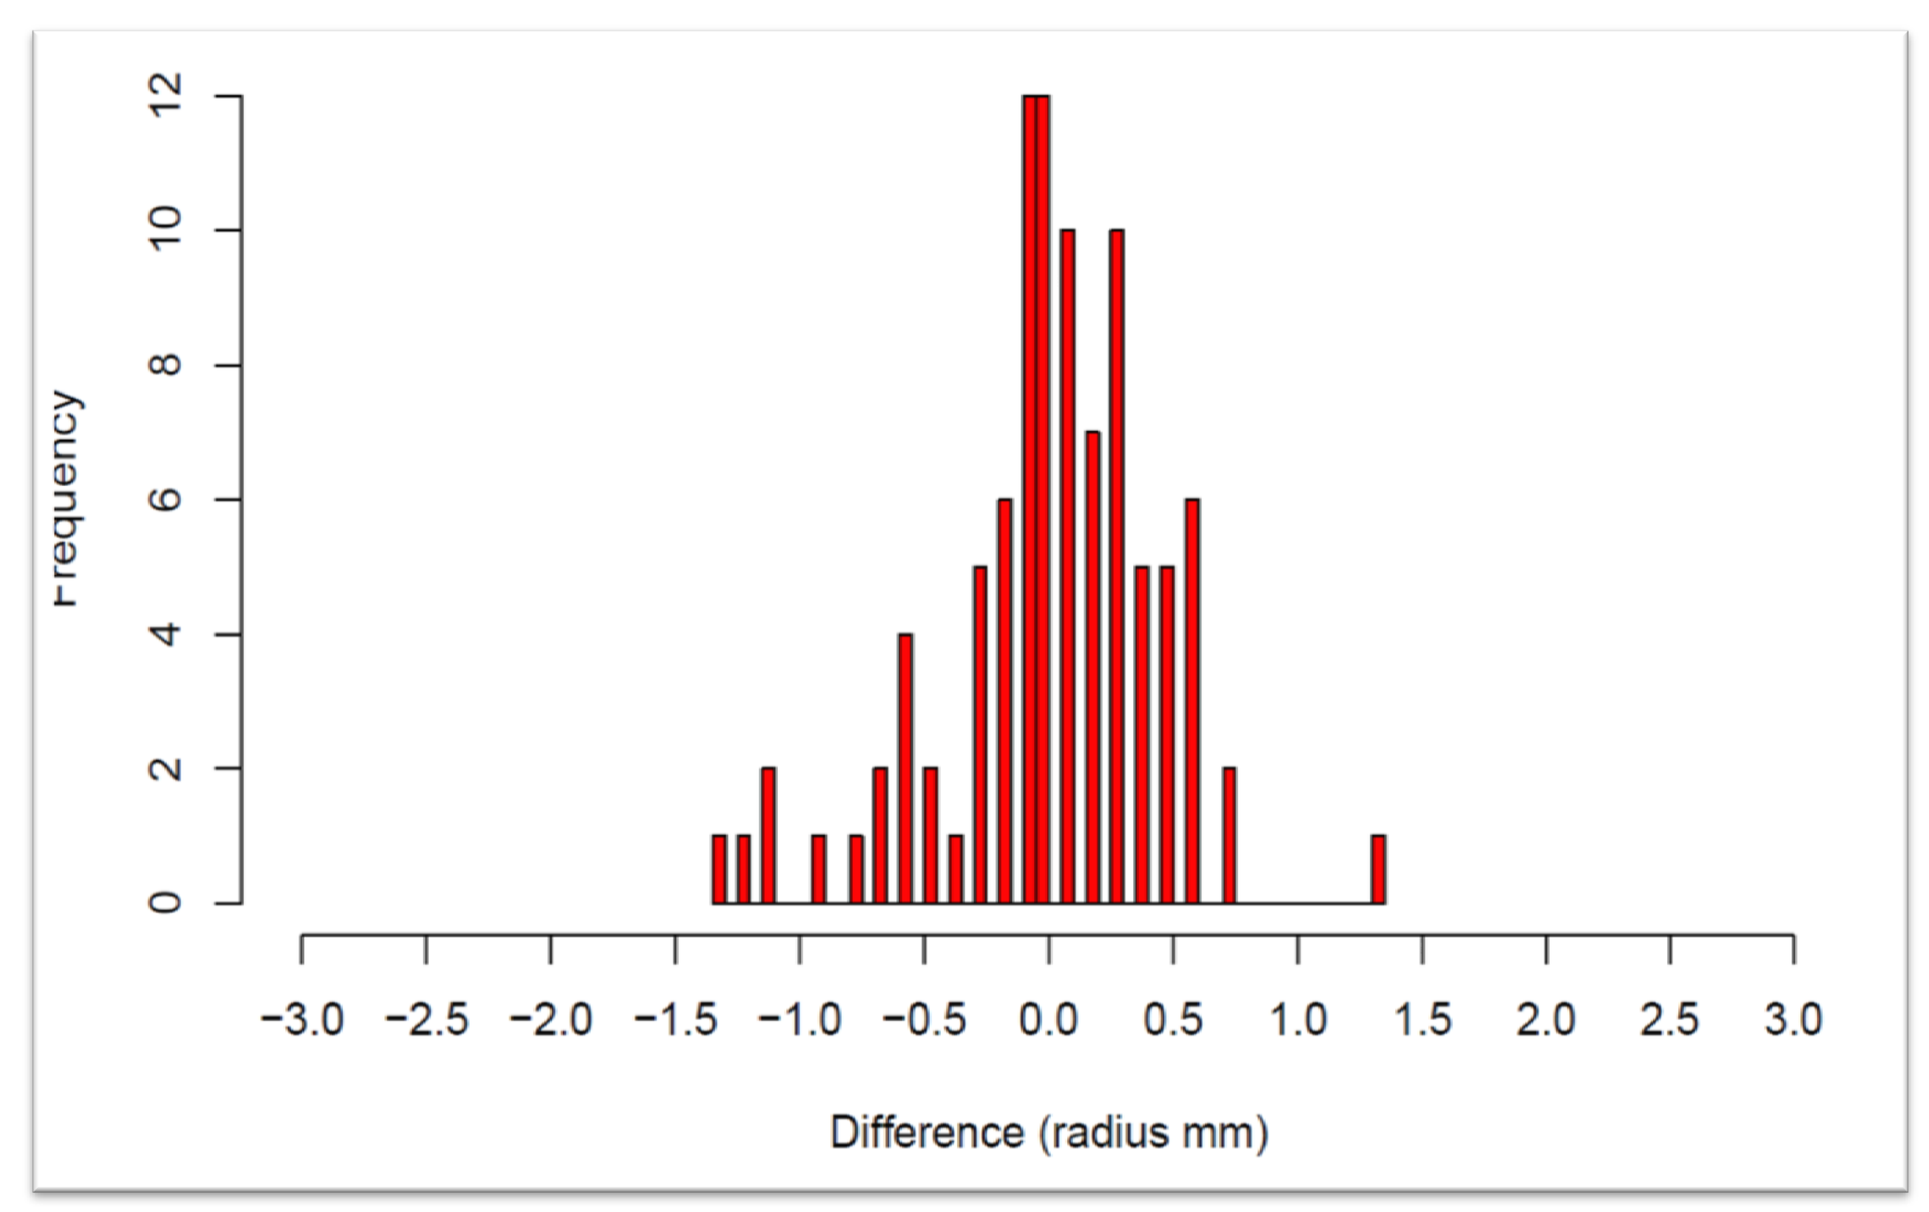
Frequency
Difference (radius mm)

Accepted:	76 of 76 zones
Correlation:	0.93

Accepted:	100 of 100 zones
Correlation:	0.99

Accepted:	96 of 152 zones
Correlation:	0.98

Conclusions

An end-user trainable machine vision system for measuring zones is presented and compared to manual zone measurements.

The proposed method shows excellent correlation between manually and automatically measured zones.

In low contrast image environments the system adapts by automatically rejecting more zones maintaining high Pearson correlation.

BD Kiestra™

NHL RESEARCH AND BUSINESS

ECCMID 2013